Predicting CD8+ cell density and tumor-immune phenotypes in non-small-cell lung cancer (NSCLC) from standard H&E slides using deep learning (DL)

David Soong[1], Becky Arbiv[2], Ettai Markovits[2], Alon Groisman[2], Yuval Shachaf[2], Tomer Dicker[2], Hisham Hamadeh[1], Kate Sasser[1], Gali Golan[2], Brandon Higgs[1], Suzana Couto[1], Ori Zelichov[2]

1: Genmab, Princeton, NJ; 2: NucleAI, Tel Aviv, Israel

Background

- Tumor infiltrated lymphocytes (TIL), namely CD8+ TILs play a major role in antitumor immunity and tumor cell eradication.
- High-density infiltration of CD8+ cells in the tumor, in contrast to CD8+ cell excluded regions, is associated with improved prognosis and response to immunotherapy in multiple cancer types.
- However, since various T-cells are indistinguishable using H&E, CD8 evaluations require additional tissue slides for IHC staining and are not performed routinely in clinical practice.
- Here, we developed DL models and derived human interpretable spatial features to predict CD8+ cell density and immune phenotypes from standard H&E slides.

Methods and materials

- 188 pairs of sequential H&E and CD8 slides from 103 patients with metastatic NSCLC were procured.
- Two board certified pathologists classified IHC slides into immune phenotypes: inflamed, desert and excluded, based on CD8 density (Tables 1, 2)
- DL models were trained to classify tumor cells, lymphocytes, fibroblasts, and tumor versus stromal areas on H&E, as well as positivity of CD8 per cell by IHC.
- 354 spatial features were calculated from the H&E slides and CD8+ density in the whole tumor region was calculated from IHC slides.
- Our biological hypotheses are based on prior knowledge from scientific literature regarding immune and spatial determinants for ICI response or immune phenotype. For example, high level of infiltration of TILs or CD8+ is correlated with response, while presence of tumor associated macrophages may confer resistance to ICI. Dozens of known immune features were screened, selected and calculated.
- Training (n=143) and test (n=45) cohorts were created and linear regression modeling predicted CD8 density from spatial H&E features.
- A multinomial logistic regression model was trained to predict these immune phenotypes from the H&E images.

Table 1.

Immune phenotype	Criteria
Inflamed	Density of CD8-positive lymphocytes in the tumor cell area >5%
Excluded	CD8+ lymphocyte presence in >50% of the invasive margin in a non-inflamed slide
Desert	Criteria of inflamed or excluded do not apply

Table 2.

Class	# of CD8 slides
Immune desert	36
Immune inflamed	70
Immune excluded	73
Borderline	19
Unusable	6

1. CD8 prediction

DL models accurately detects various cell types and classifies regions into tumor vs stroma on H&E images. Various areas including tumor core, tumor invasive margin, adjacent tumor microenvironment, outer tumor microenvironment, were derived for spatial feature calculation.

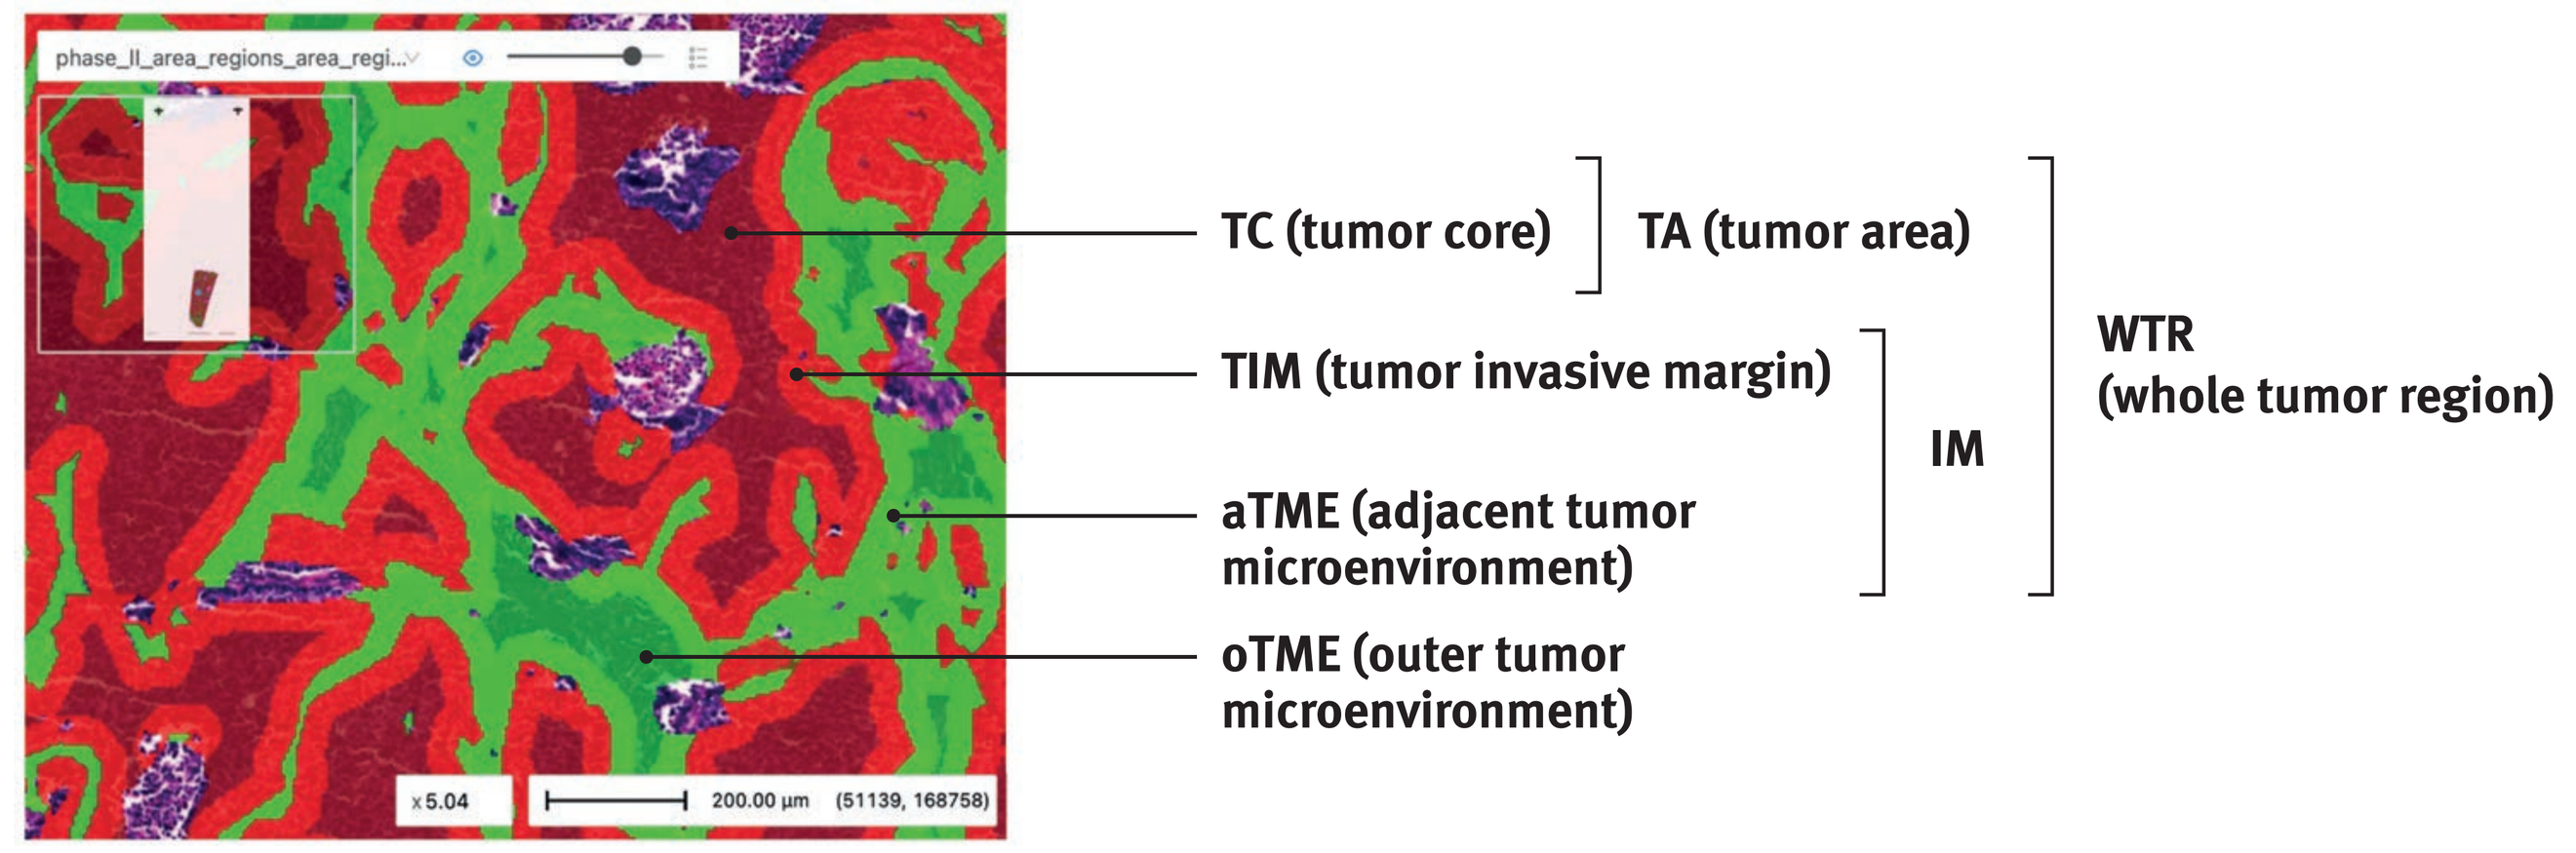
TC (tumor core)
TA (tumor area)
TIM (tumor invasive margin)
IM
WTR
(whole tumor region)
aTME (adjacent tumor microenvironment)
oTME (outer tumor microenvironment)
x5.04
200.00 µm (51139, 168758)

Figure 1: Areas defined in the tumor microenvironment. A fixed distance of 40 µm from the tumor area edge was used to calculate the TIM and the aTME, while the oTME is defined as the stromal area found between the aTME and 500 µm from the tumor edge into the stroma.

Predicted CD8 density was highly correlated with pathologist's estimates on the test set ($r = 0.87$; $p<0.0001$). The H&E features most predictive of CD8 density were related to lymphocyte densities, tumor-lymphocyte proximity in the invasive margin, and lymphocyte density in the tumor area.

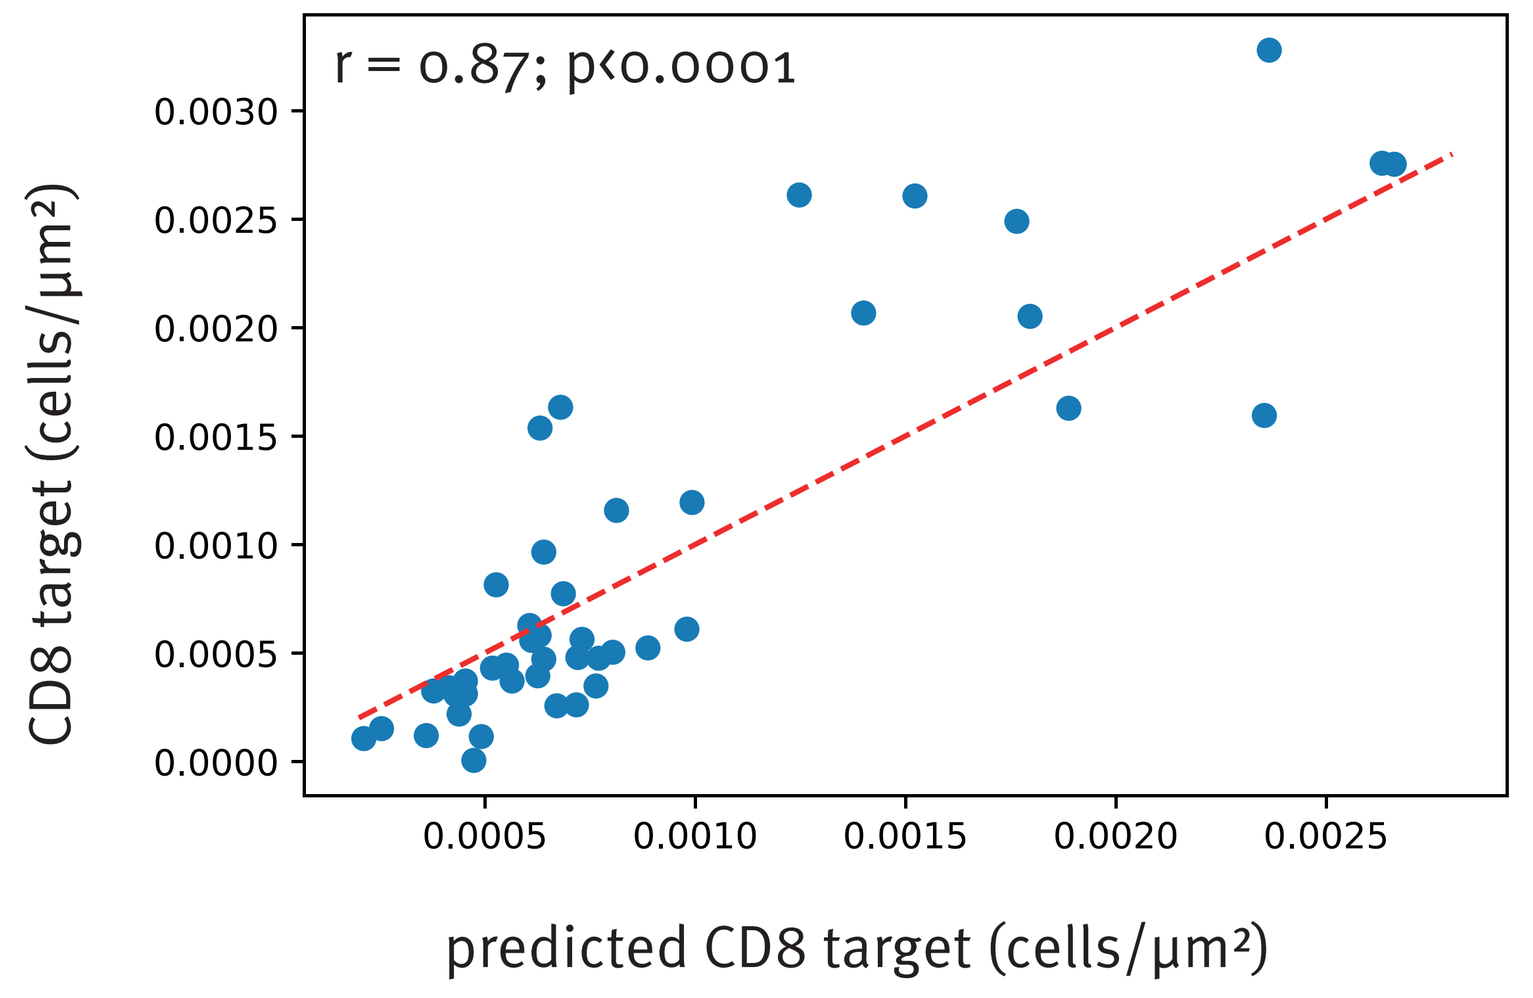
r = 0.87; p<0.0001
CD8 target (cells/µm²)
0.0030
0.0025
0.0020
0.0015
0.0010
0.0005
0.0000
0.0005
0.0010
0.0015
0.0020
0.0025
predicted CD8 target (cells/µm²)

Figure 2: Correlation between predicted CD8 density and pathologist's estimates.

2. Immune phenotype prediction

The H&E features most predictive of immune phenotype were proximity between lymphocytes and tumor cells or granulocytes, as well as lymphocyte:fibroblast density ratio.
Manual review of predictions by pathologists on the test set images demonstrated an accuracy of 74% (95% CI 57%-85%, $p<0.0001$) in classifying slides to the correct immune phenotypes (Table 3).

Table 3. Immune phenotype classification

Prediction	Reference: Desert	Reference: Exclude	Reference: Inflamed
Desert	6	2	1
Exclude	0	12	4
Inflamed	1	3	12

3. Phenotype visualisation examples

Inflamed

Excluded

Desert

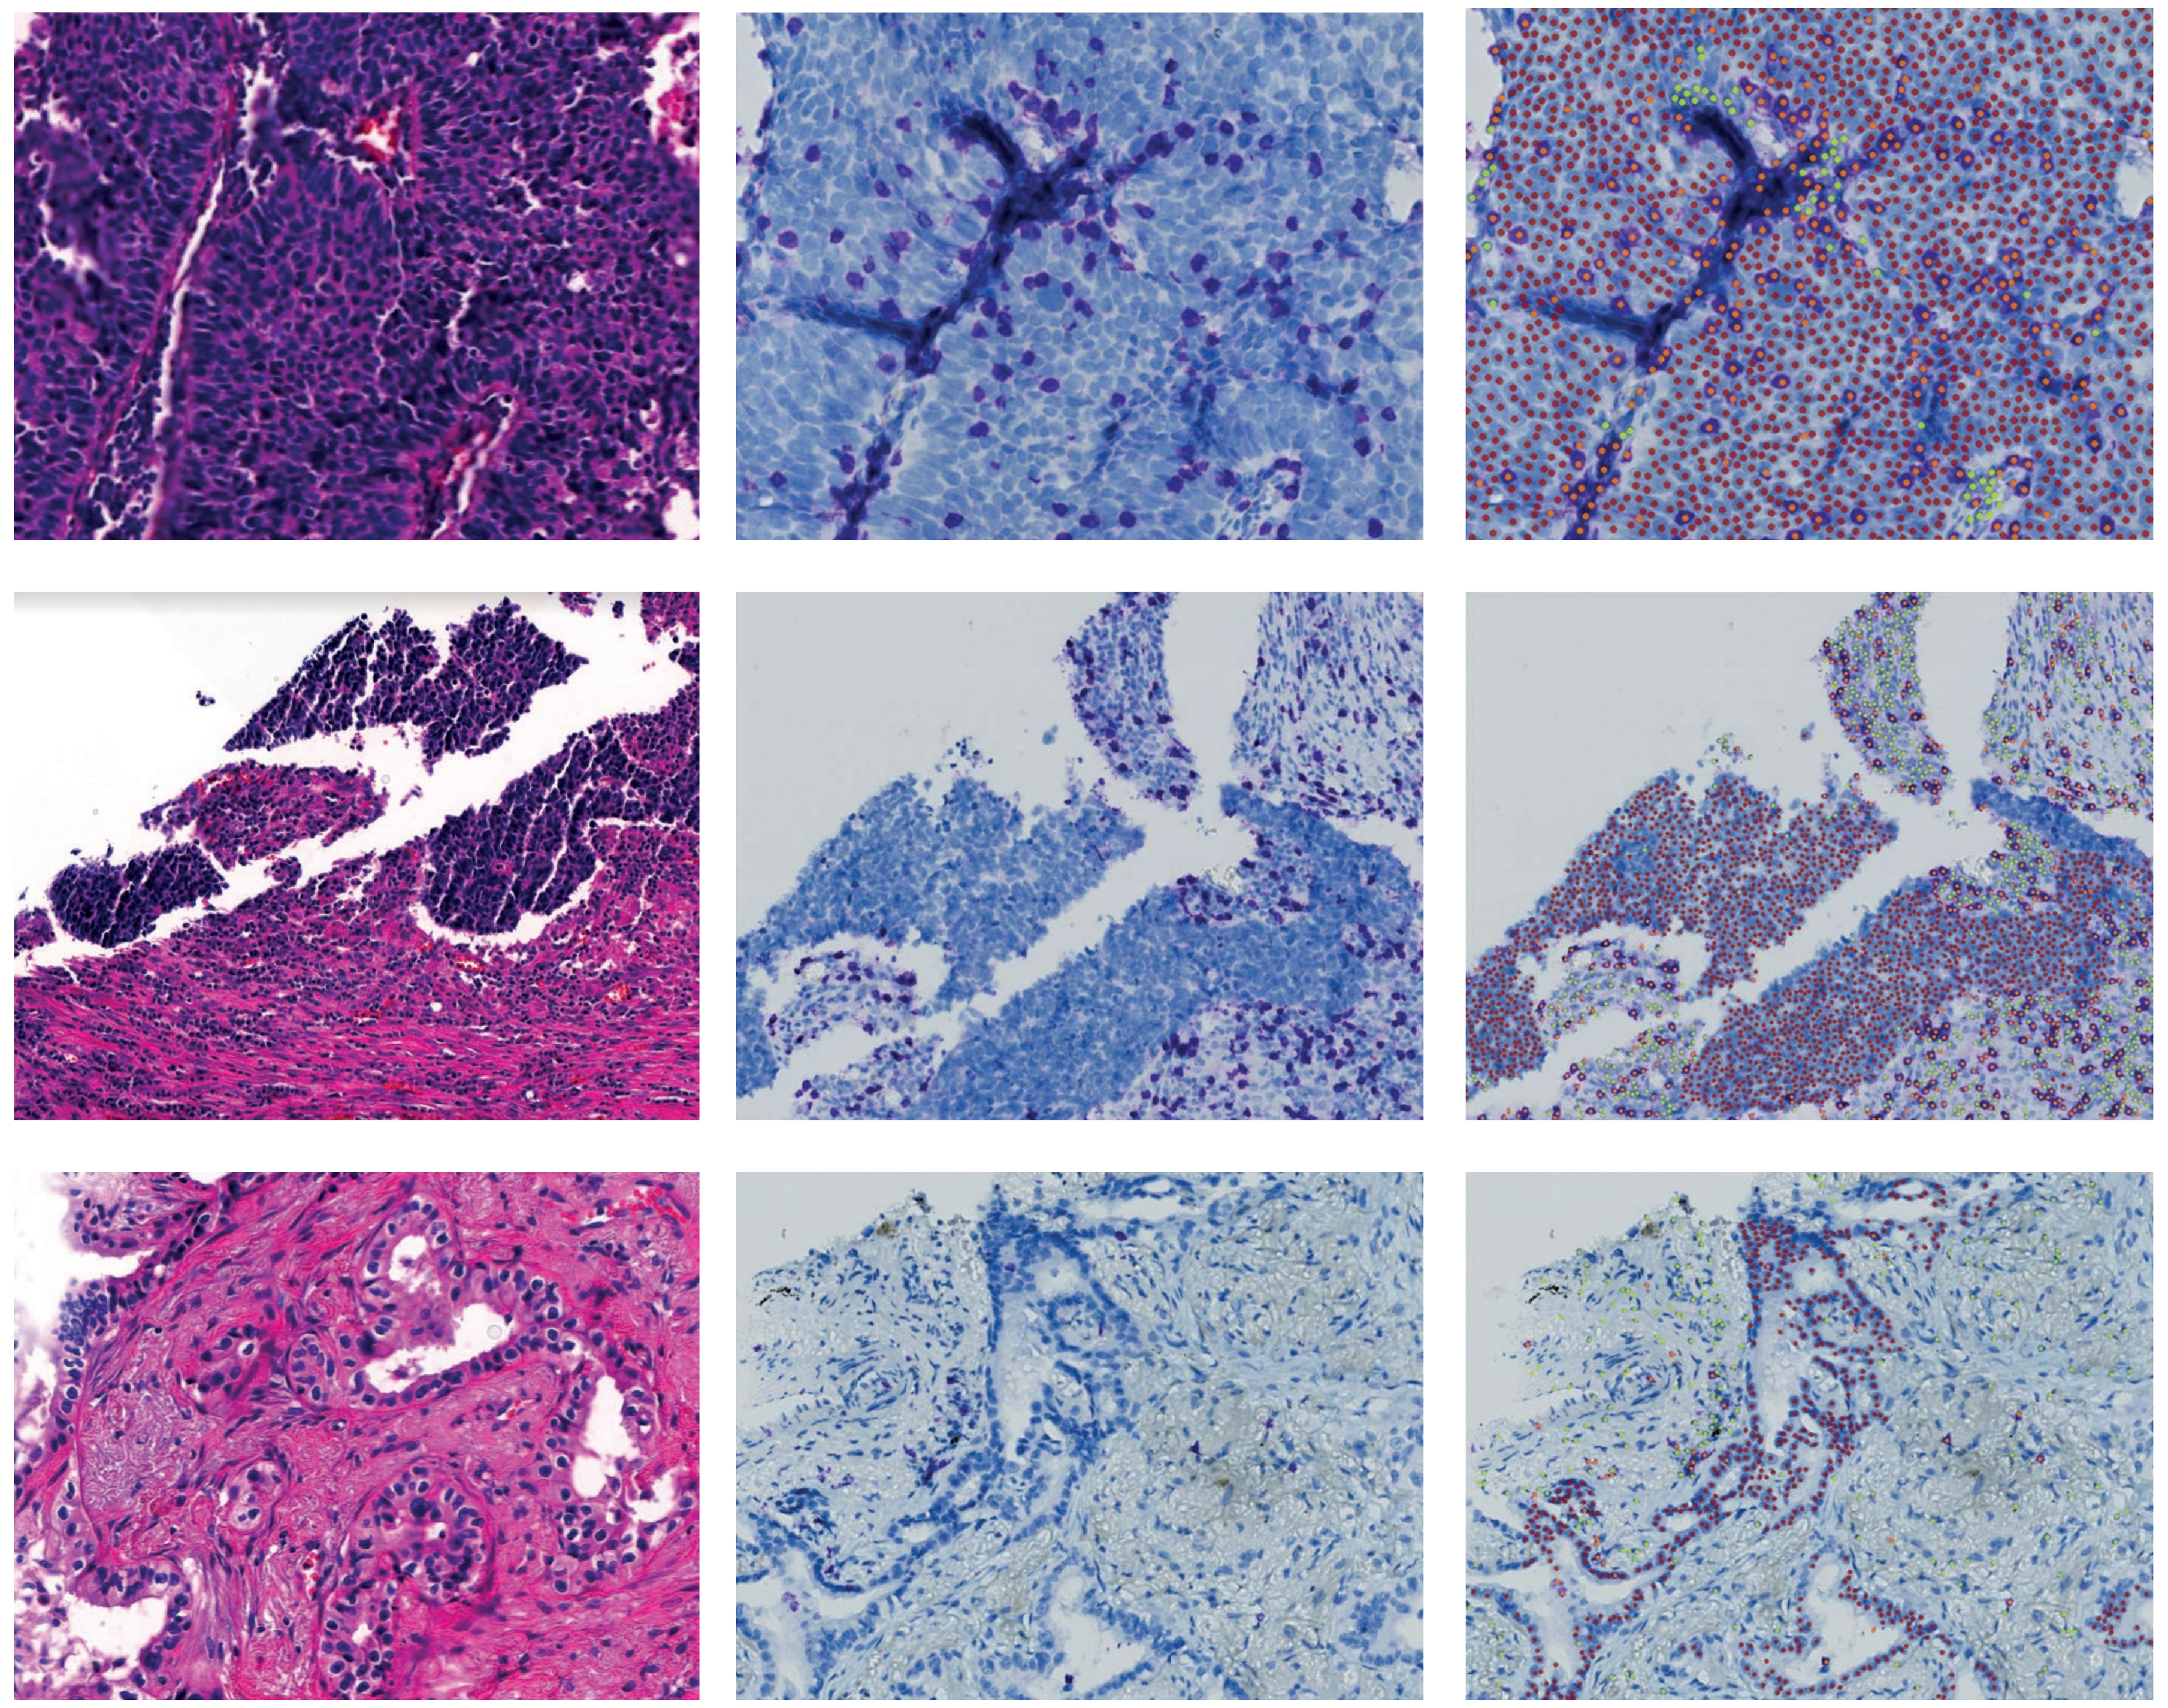

Figure 3. Areas defined in the tumor microenvironment. A fixed distance of 40 µm from the tumor area edge was used to calculate the TIM and the aTME, while the oTME is defined as the stromal area found between the aTME and 500 µm from the tumor edge into the stroma.

Left side: raw H&E slide. Middle: raw CD8. Right side: cell detection algorithm results (red - tumor cells, yellow - CD8-negative, orange - CD8-positive).

Conclusions

- Spatial analysis of immune and tumor cells from standard H&E slides using DL identified human interpretable features that accurately predicted CD8 cell density and identified immune phenotypes.
- Our framework is easily generalizable and can be extended to predict other IHC stains or gene signatures.
- By only requiring H&E images, these biomarkers important for checkpoint inhibitor therapy can be measured in clinical practice without the requirement of IHC staining.

References

- Kovacsovics-Bankowski et al. J Immunother Cancer. 2014; 2: 38.
- Li et al. eClinicalMedicine 2021; 41, 101134
- El Bairi et al. npj Breast Cancer 2021; 7, 150

Society for Immunotherapy of Cancer 37th Annual Meeting, November 8-12, 2022